The 96th annual meeting of American Association of Cancer Research. April 16~20th, 2005, at Anaheim CA, USA

A novel antitumor effect of water-extract from *Pleurotus cornucopiae* (Tamogitake; Golden Oyster Mushroom) that promotes dendritic cell activation and down-regulation of regulatory T cells in tumor-bearing mice

Kazunori Kato[1], Shinobu Kaihou[2], Shin-ya Hagiwara[3], Takahiro Tomiyama[2] and Hirofumi Hamada[1]

[1]Department of Molecular Medicine, Sapporo Medical University, [2]R&D Department, Three B Co. Inc., [3]Division of Environmental Resources, Hokkaido University, Sapporo, Japan

Abstract

The fruit body of the mushroom *Pleurotus cornucopiae* (Tamogitake; Golden Oyster Mushroom) has traditionally been used as a health food for the prevention of hypertension, diabetes and cancer. In the present study, we examined for the antitumor activity of the extract of *Pleurotus cornucopiae* using a sarcoma 180 (S180) transplantation experimental model mice and its immune adjuvant mechanism on dendritic cells (DC) and regulatory T cells (T-reg).

ICR mice were transplanted intramuscular injection with S180 (5 million cells/mouse) followed by oral administration of hot water extract of *Pleurotus cornucopiae* at a dose of 4.0 ml/kg/day for 21 or 35 days. The administration of *Pleurotus cornucopiae* resulted in significant inhibition of S180 tumor growth and prolonged the survival compared with saline-administrated tumor-bearing mice. In order to clarify the antitumor mechanism of *Pleurotus cornucopiae*, we investigated the immuno-modulatory effect on the activation of DC *in vitro*. In contrast to immature DC, treatment of DC with *Pleurotus cornucopiae* extract expressed high levels of costimulatory molecules (i.e., CD54 and CD86), and activation/maturation markers (i.e., CD25 and CD83). Interleukin 12 (IL-12) is produced in large amounts by mature, not by immature, DC and therefore is a suitable factor to determine DC maturation. DC stimulated with *Pleurotus cornucopiae* extract could produce significantly higher levels of IL-12 in a dose-dependent manner than those stimulated without or with *Agaricus blazei*.

We next investigated the possibility that T-reg populations may be responsible for the antitumor effect of *Pleurotus cornucopiae*. Consistent with antitumor effect, sarcoma-bearing ICR mice orally administrated with the extract of *Pleurotus cornucopiae* have significantly decreased numbers of $CD4^+GITR^+$ T-reg population in spleen (13.2 ± 3.1%, n=6) compared with saline-treated mice (33.4 ± 10.6%, n=6). We also found that 3LL lung tumor-bearing mice administrated with *Pleurotus cornucopiae* have significantly decreased numbers of $CD4^+CD25^+$ T-reg population in spleen (16.2 ± 0.6% vs. 24.7 ± 3.8% in saline-treated mice, n=5). Collectively, these findings indicate that *Pleurotus cornucopiae* contains active substances that display its ability to enhance antitumor immunity mediated by the activation of DC and down-regulation of T-reg.

Experimental Design

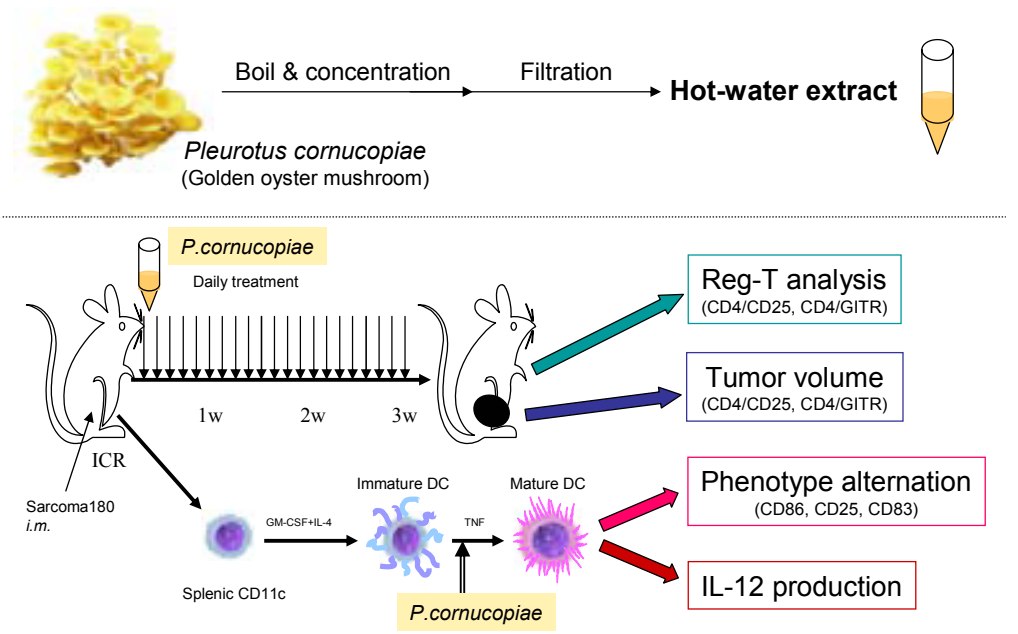
Boil & concentration
Filtration
Hot-water extract
Pleurotus cornucopiae
(Golden oyster mushroom)
P.cornucopiae
Daily treatment
1w
2w
3w
ICR
Sarcoma180
i.m.
Splenic CD11c
GM-CSF+IL-4
Immature DC
TNF
Mature DC
P.cornucopiae
Reg-T analysis
(CD4/CD25, CD4/GITR)
Tumor volume
(CD4/CD25, CD4/GITR)
Phenotype alternation
(CD86, CD25, CD83)
IL-12 production

Summary

- Orally administration of water-extract of *P.cornucopiae* inhibited the growth of Sarcoma 180 in ICR mice.
- *P.cornucopiae* induced costimulatory molecule expression and IL-12 secretion by dendritic cells.
- CD25/GITR-positive regulatory T cells were decreased in tumor-bearing mice administrated with *P.cornucopiae*.
- In conclusion, *Pleurotus cornucopiae* contains substance(s) that has potent antitumor activity and a novel immune-modulatory activity.

Materials and Methods

<u>Natural materials and preparation of water-extract from *Pleurotus cornucopiae*.</u>

Hot-water extract of *P. cornucopiae* (golden oyster mushroom) cultured artificially in Three B Co., Inc. (Hokkaido, Japan) was prepared as follows: the fruit bodies of *P. cornucopiae* (350 g) were directly extracted with distilled water (200 ml) at 94° C for 10 min. After concentration to 80 ml with evaporator, the insoluble residue was removed through the filtration of 1.0µm and 0.2µm, subsequently, and the water-extract solution was stored in 4° C before use.

<u>Tumor</u>

Sarcoma 180 was supplied by Tohoku University, Bioresource Center (Sendai, Japan), and maintained by intraperitoneal passages in ICR mice (SLC Inc., Shizuoka, Japan). Lewis lung carcinoma LLC was supplied by RIKEN Bioresource Center (Tsukuba, Japan), and maintained *in vitro* in RPMI1640 medium supplemented with 10% FCS.

<u>Animal experiments</u>

Six-week-old female ICR mice were inoculated with Sarcoma 180 (5 x10^6 cells/mouse) *subcutaneously* in the left thigh of each mouse, and Six-week-old female C57BL/6 mice were inoculated with LLC (1 x10^6 cells/mouse) *subcutaneously* in the dorsal area of each mouse. Each tumor-inoculated mice were divided into 2 groups for saline-treated or *P. cornucopiae*-treated group. Each group of 6 mice was orally administrated with saline or water-extract of *P. cornucopiae* daily at 4 ml/kg. The tumor diameter was measured using digital calipers (Mitutoyo Co., Kawasaki, Japan) and the volume was calculated as follows: *tumor vol. (mm^3) = 0.4* x *the long diameter (mm)* x *the short diameter (mm)* x *the short diameter (mm)*. Body weights were recorded on each day. On day 22 or 29, mice were anesthetized and sacrificed; the spleen was removed and used for flow cytometry.

<u>Stimulation of dendritic cells</u>

C57BL/6 or ICR mice spleen were minced and digested in 1 mg/ml collagenase in HBSS for 30 min at 37° C. Cell suspension was filtered through nylon mesh (40µm) and washed with RPMI1640 medium. Cells were then incubated with mouse CD11c-microbeads following manufacturer's instructions (Miltenyi Biotec GmbH, Germany). Positive selection was performed using AutoMACS, and enriched dendritic cell (DC) population was used in this study. $CD11c^+$ DCs were cultured in RPMI1640 medium containing 10% FCS and GM-CSF (500 U/ml) and IL-4 (50 ng/ml) for 4 days. Cultured DCs were harvested and adjusted to the concentration of 2 x 10^5 cells/ml in RPMI1640 medium with or without TNFα (50 ng/ml), and treated with water-extract of *P. cornucopiae* at 0.3%, 0.1% or 0.03% *(v/v)*. As control, we also used water-extract of *Agaricus blazei* as same concentration (prepared in Three B Inc.). After 24 h, stimulated DCs were harvested and tested for phenotypical alternation by flow cytometory using specific mAbs to CD86, CD25 and CD83 (eBioscience, San Diego, CA). Cell-free supernatant was also harvested and tested for the production of IL-12 by ELISA (R&D Systems Inc., Minneapolis, MN).

<u>Analysis of regulatory T cells in tumor-bearing mice</u>

Splenocytes isolated from tumor-bearing mice that were administrated with saline or water-extract of *P. cornucopiae* daily were incubated with specific mAbs for CD4 (FITC), CD25 (PE) or GITR (PE). The labeled cells were analyzed via flow cytometry using a FACSCalibur (BD, San Jose, CA). The percentage of regulatory T cells were calculated by gating $CD4^+/CD25^+$ or $CD4^+/GITR^+$ cells using FlowJo software (Tree Star Inc., Ashland, OR).

Results

Effect of oral administration of water-extract of *P.cornucopiae* on sarcoma-180 growth in ICR mice

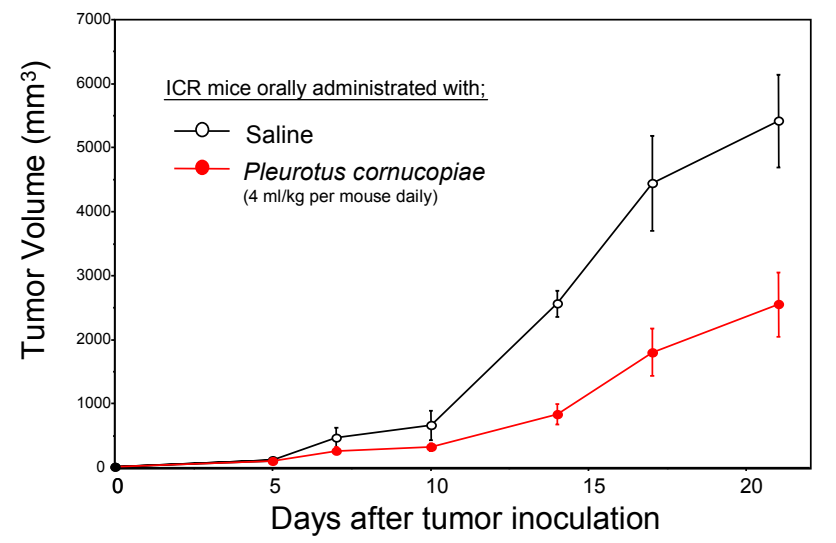
Tumor Volume (mm³)
ICR mice orally administrated with;
Saline
Pleurotus cornucopiae
(4 ml/kg per mouse daily)
Days after tumor inoculation
0
1000
2000
3000
4000
5000
6000
7000
0
5
10
15
20

Induction of immune accessory molecules in DC activation and maturation by *P.cornucopiae in vitro*

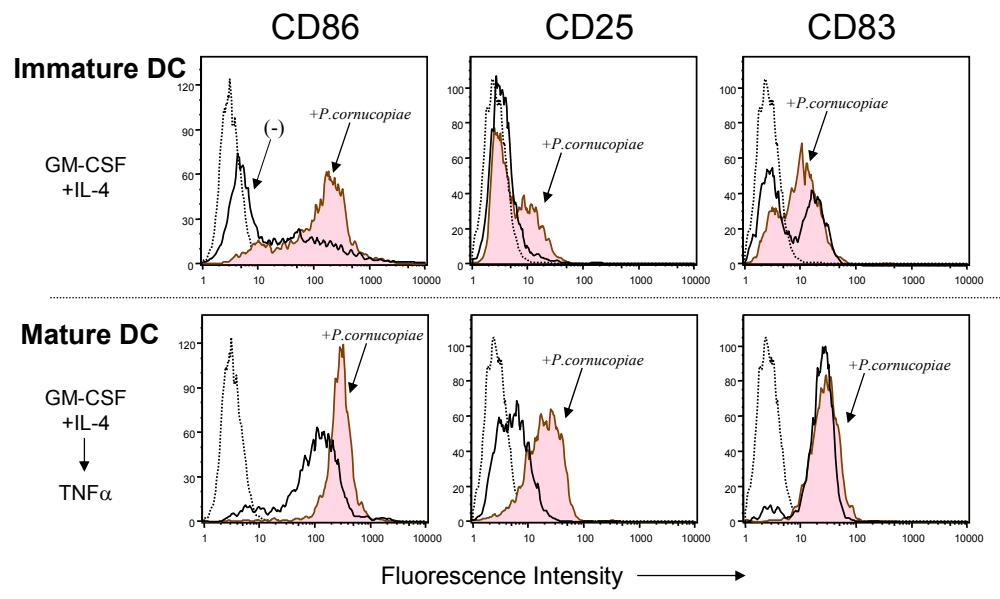
CD86
CD25
CD83
Immature DC
GM-CSF
+IL-4
Mature DC
GM-CSF
+IL-4
TNFα
(-)
+P.cornucopiae
Fluorescence Intensity

Increased production of IL-12 p70 by dendritic cells stimulated with *P.cornucopiae in vitro*

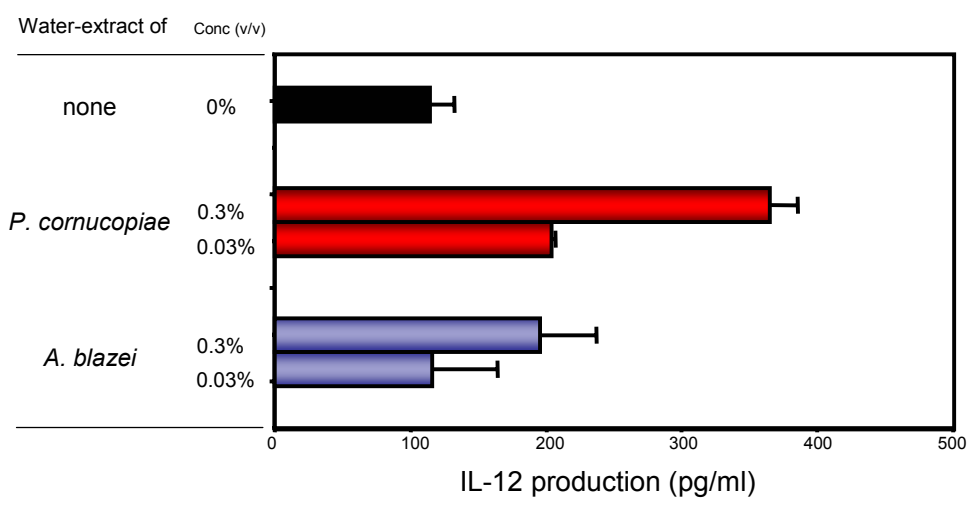
Water-extract of
Conc (v/v)
none
0%
P. cornucopiae
0.3%
0.03%
A. blazei
0.3%
0.03%
0
100
200
300
400
500
IL-12 production (pg/ml)

Aggregation and morphological changes of dendritic cells stimulated with *P.cornucopiae in vitro*

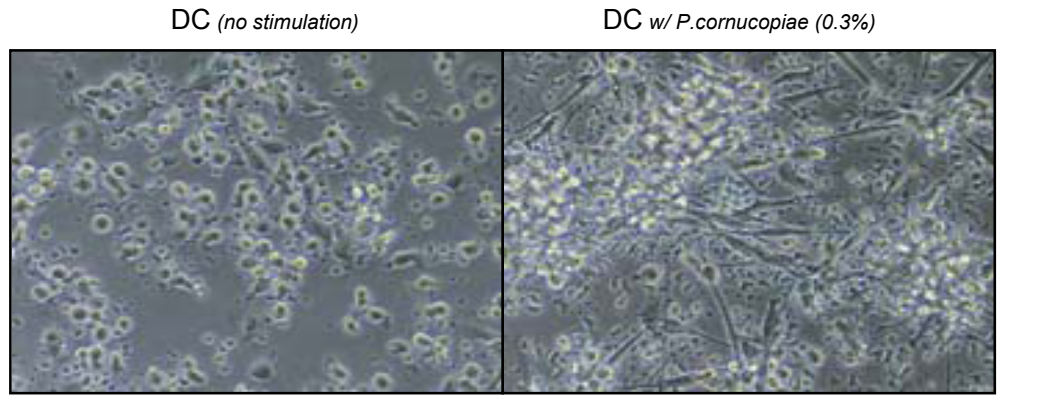
DC (no stimulation)
DC w/ P.cornucopiae (0.3%)

Oral administration of *P.cornucopiae* suppress regulatory T cells in sarcoma-bearing ICR mice

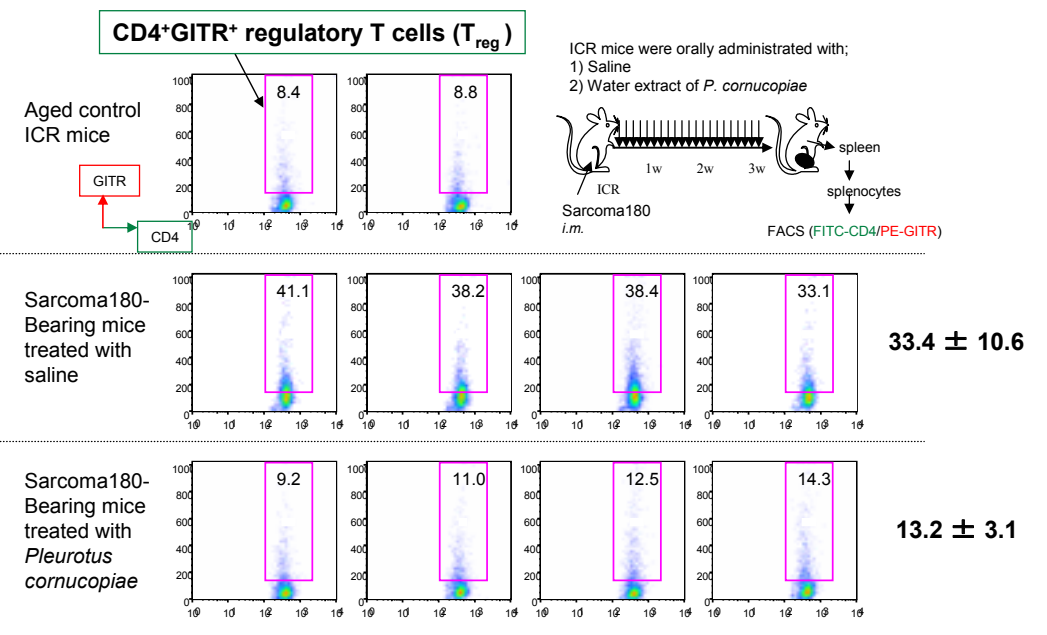
CD4⁺GITR⁺ regulatory T cells (Treg)
ICR mice were orally administrated with;
1) Saline
2) Water extract of P. cornucopiae
ICR
Sarcoma180
i.m.
1w
2w
3w
spleen
splenocytes
FACS (FITC-CD4/PE-GITR)
Aged control
ICR mice
GITR
CD4
8.4
8.8
Sarcoma180-
Bearing mice
treated with
saline
41.1
38.2
38.4
33.1
33.4 ± 10.6
Sarcoma180-
Bearing mice
treated with
Pleurotus
cornucopiae
9.2
11.0
12.5
14.3
13.2 ± 3.1

Oral administration of *P.cornucopiae* suppress regulatory T cells in LLC-bearing C57BL/6 mice

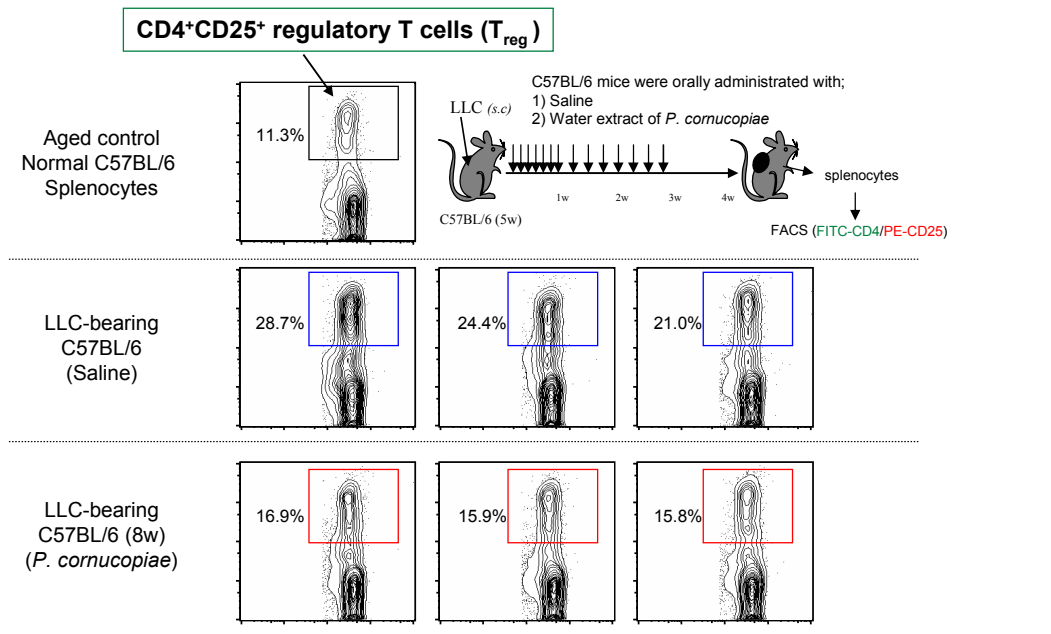
CD4⁺CD25⁺ regulatory T cells (Treg)
C57BL/6 mice were orally administrated with;
1) Saline
2) Water extract of P. cornucopiae
LLC (s.c)
C57BL/6 (5w)
1w
2w
3w
4w
splenocytes
FACS (FITC-CD4/PE-CD25)
Aged control
Normal C57BL/6
Splenocytes
11.3%
LLC-bearing
C57BL/6
(Saline)
28.7%
24.4%
21.0%
LLC-bearing
C57BL/6 (8w)
(P. cornucopiae)
16.9%
15.9%
15.8%